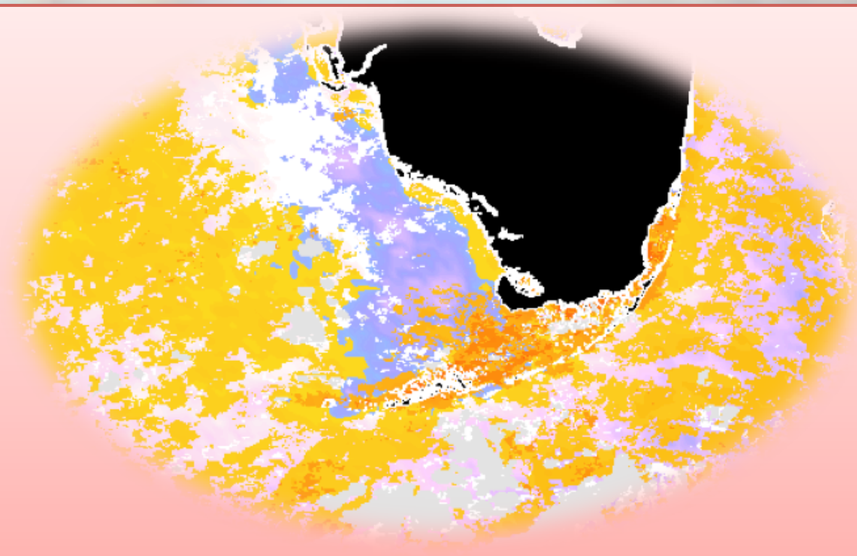

Florida Keys coral reef diversity buffered by intermediate sea surface temperature variability

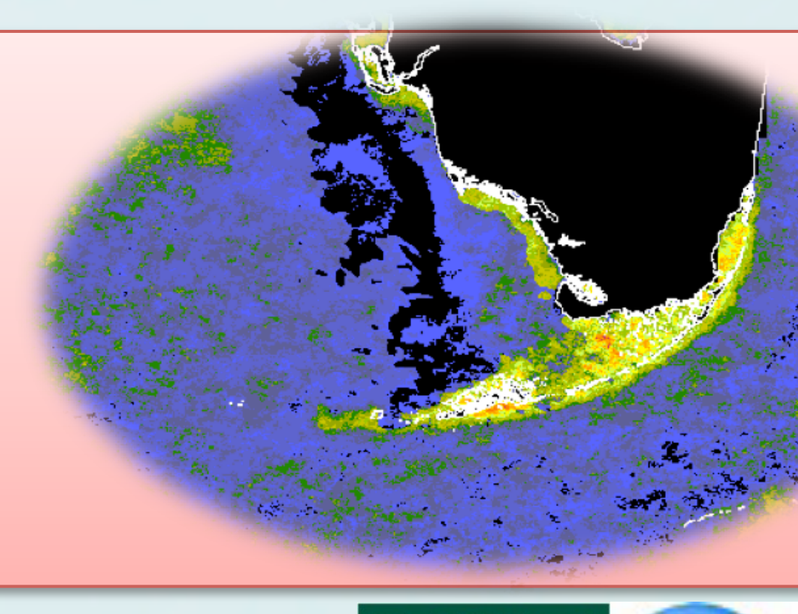

M. Vega-Rodriguez[1*], F. E. Müller-Karger[1], I. Soto[1], G. A. Quiles-Perez[1], C. M. Eakin[2], J. Li[2], L. Guild[3], S. Lynds[4], D. L. Jones[1], R. Ruzicka[5], M. Collella[5], P. Hallock[1]

Introduction

- ❖ Stony coral benthic cover in the Florida Keys National Marine Sanctuary declined on average of >50% between 1996 and 2010. While the ultimate cause for this decline remains unclear, stress due to extreme high and low water temperatures is thought to be one driving factor.

- ❖ We examined this possibility using observations of benthic cover of stony coral species, octocorals, sponges and macroalgae obtained from the CREMP surveys, and satellite-derived temperature observations.

Null Hypothesis: Sea surface temperature (SST) variability and acute warm temperature events had no influence on the coral reef diversity in the Florida Keys between 1996 and 2010.

Methods

- ❖ 33 permanent CREMP sites within the Florida Keys National Marine Sanctuary and 3 in Dry Tortugas National Marine Park were used (Figure 1).

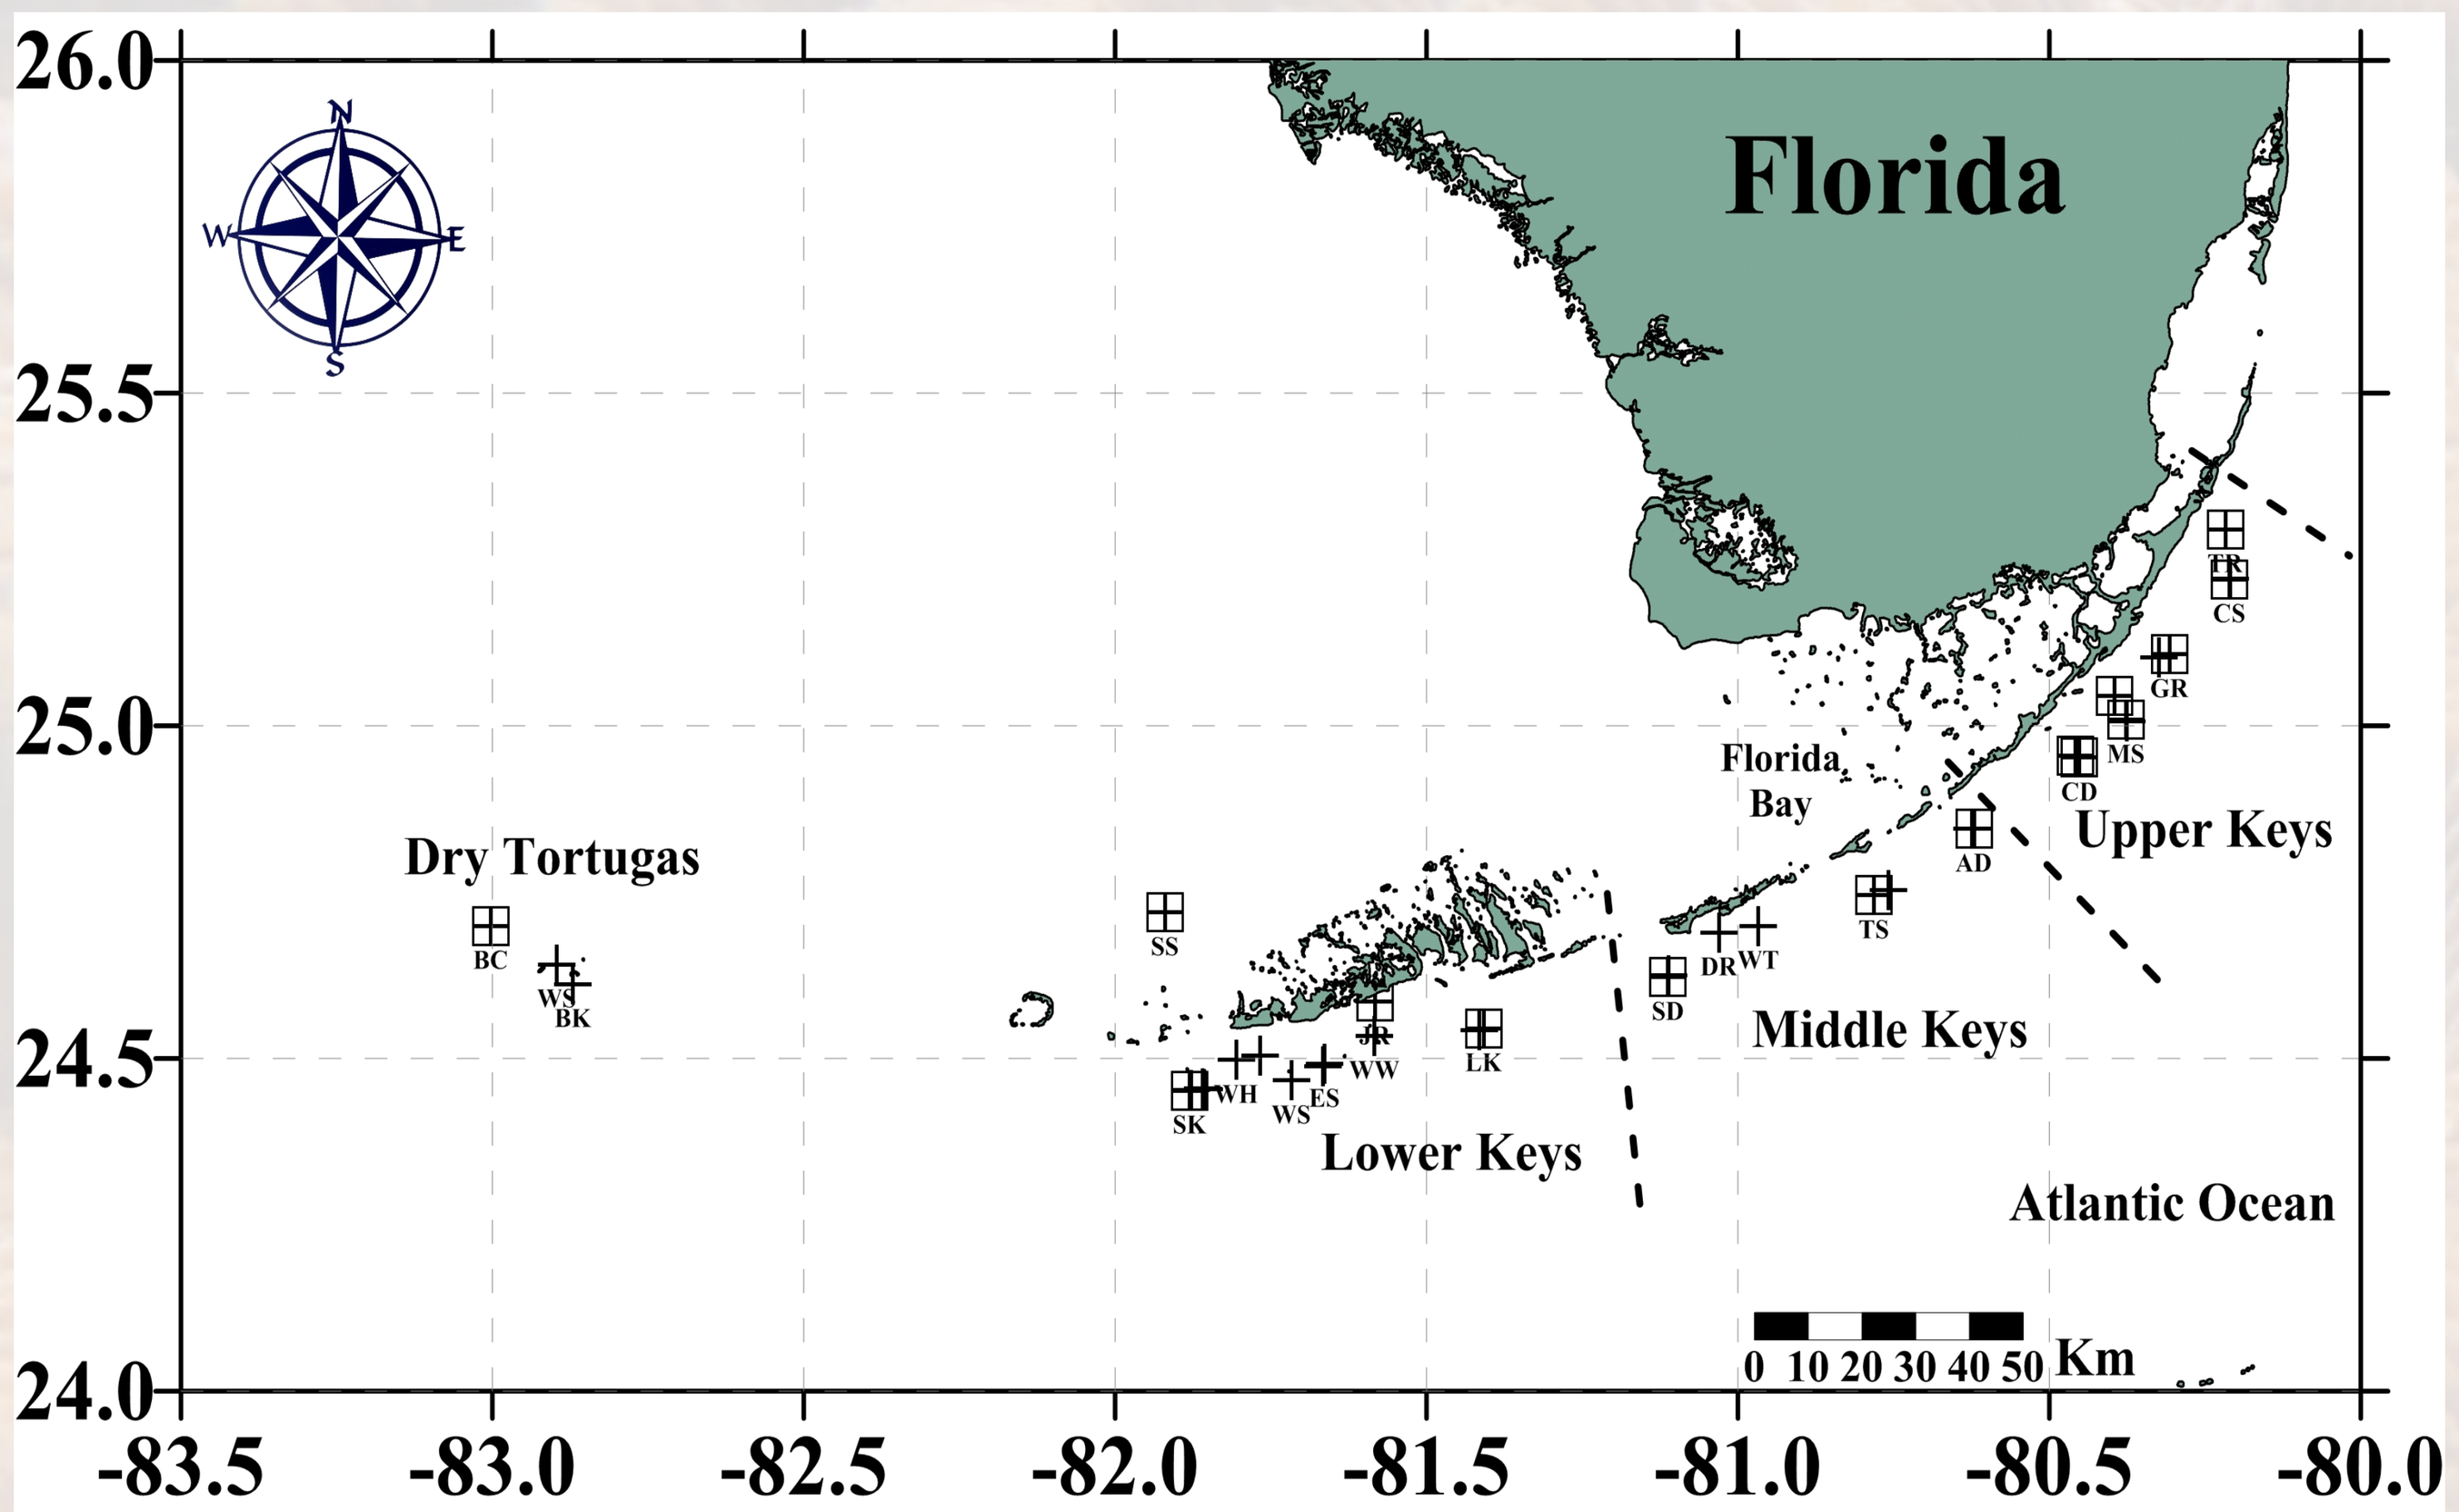

Florida
Dry Tortugas
Florida Bay
Upper Keys
Middle Keys
Lower Keys
Atlantic Ocean
0 10 20 30 40 50 Km
26.0
25.5
25.0
24.5
24.0
-83.5
-83.0
-82.5
-82.0
-81.5
-81.0
-80.5
-80.0

Figure 1. CREMP stations used in this study. Boxes with crosses represent stations for which all satellite derived SST data, *in situ* temperature data and CREMP annual surveys were available.

Regional AVHRR SST means (1 km^2) calculated from weekly means were used for obtaining:

- ❖ Annual SST variances categorized in 3 levels: low (< 7 °C), intermediate (7 – 10.9 °C) and, high (≥ 11 °C).
- ❖ Difference between summer and winter monthly regional means (Figure 2).
- ❖ High-resolution HotSpots and Degree Heating Weeks (DHW's).

Multivariate statistics used:

- ❖ np-MANOVA: to test the significance of the variation of coral species cover with respect to SST variability.
- ❖ Distance-based Canonical Analysis of Principal Coordinates (CAP) constrained ordinations: provided visualizations of coral species responsible for the multivariate dispersion respective to SST variability.

Univariate statistics used:

- ❖ Regional means for stony coral species richness were obtained.
- ❖ Regional means for Shannon diversity indices were calculated using percent cover data for each stony coral species and equation 1:

$$H' = -\sum p_i \ln p_i, \quad (1)$$

Preliminary Results

- ❖ All regions experienced the most extreme temperature differences (6.5 - 7.5 °C) between 2009 and 2010. These differences were generally the highest for Lower Keys (LK) and the Dry Tortugas (DT) (Figure 2).

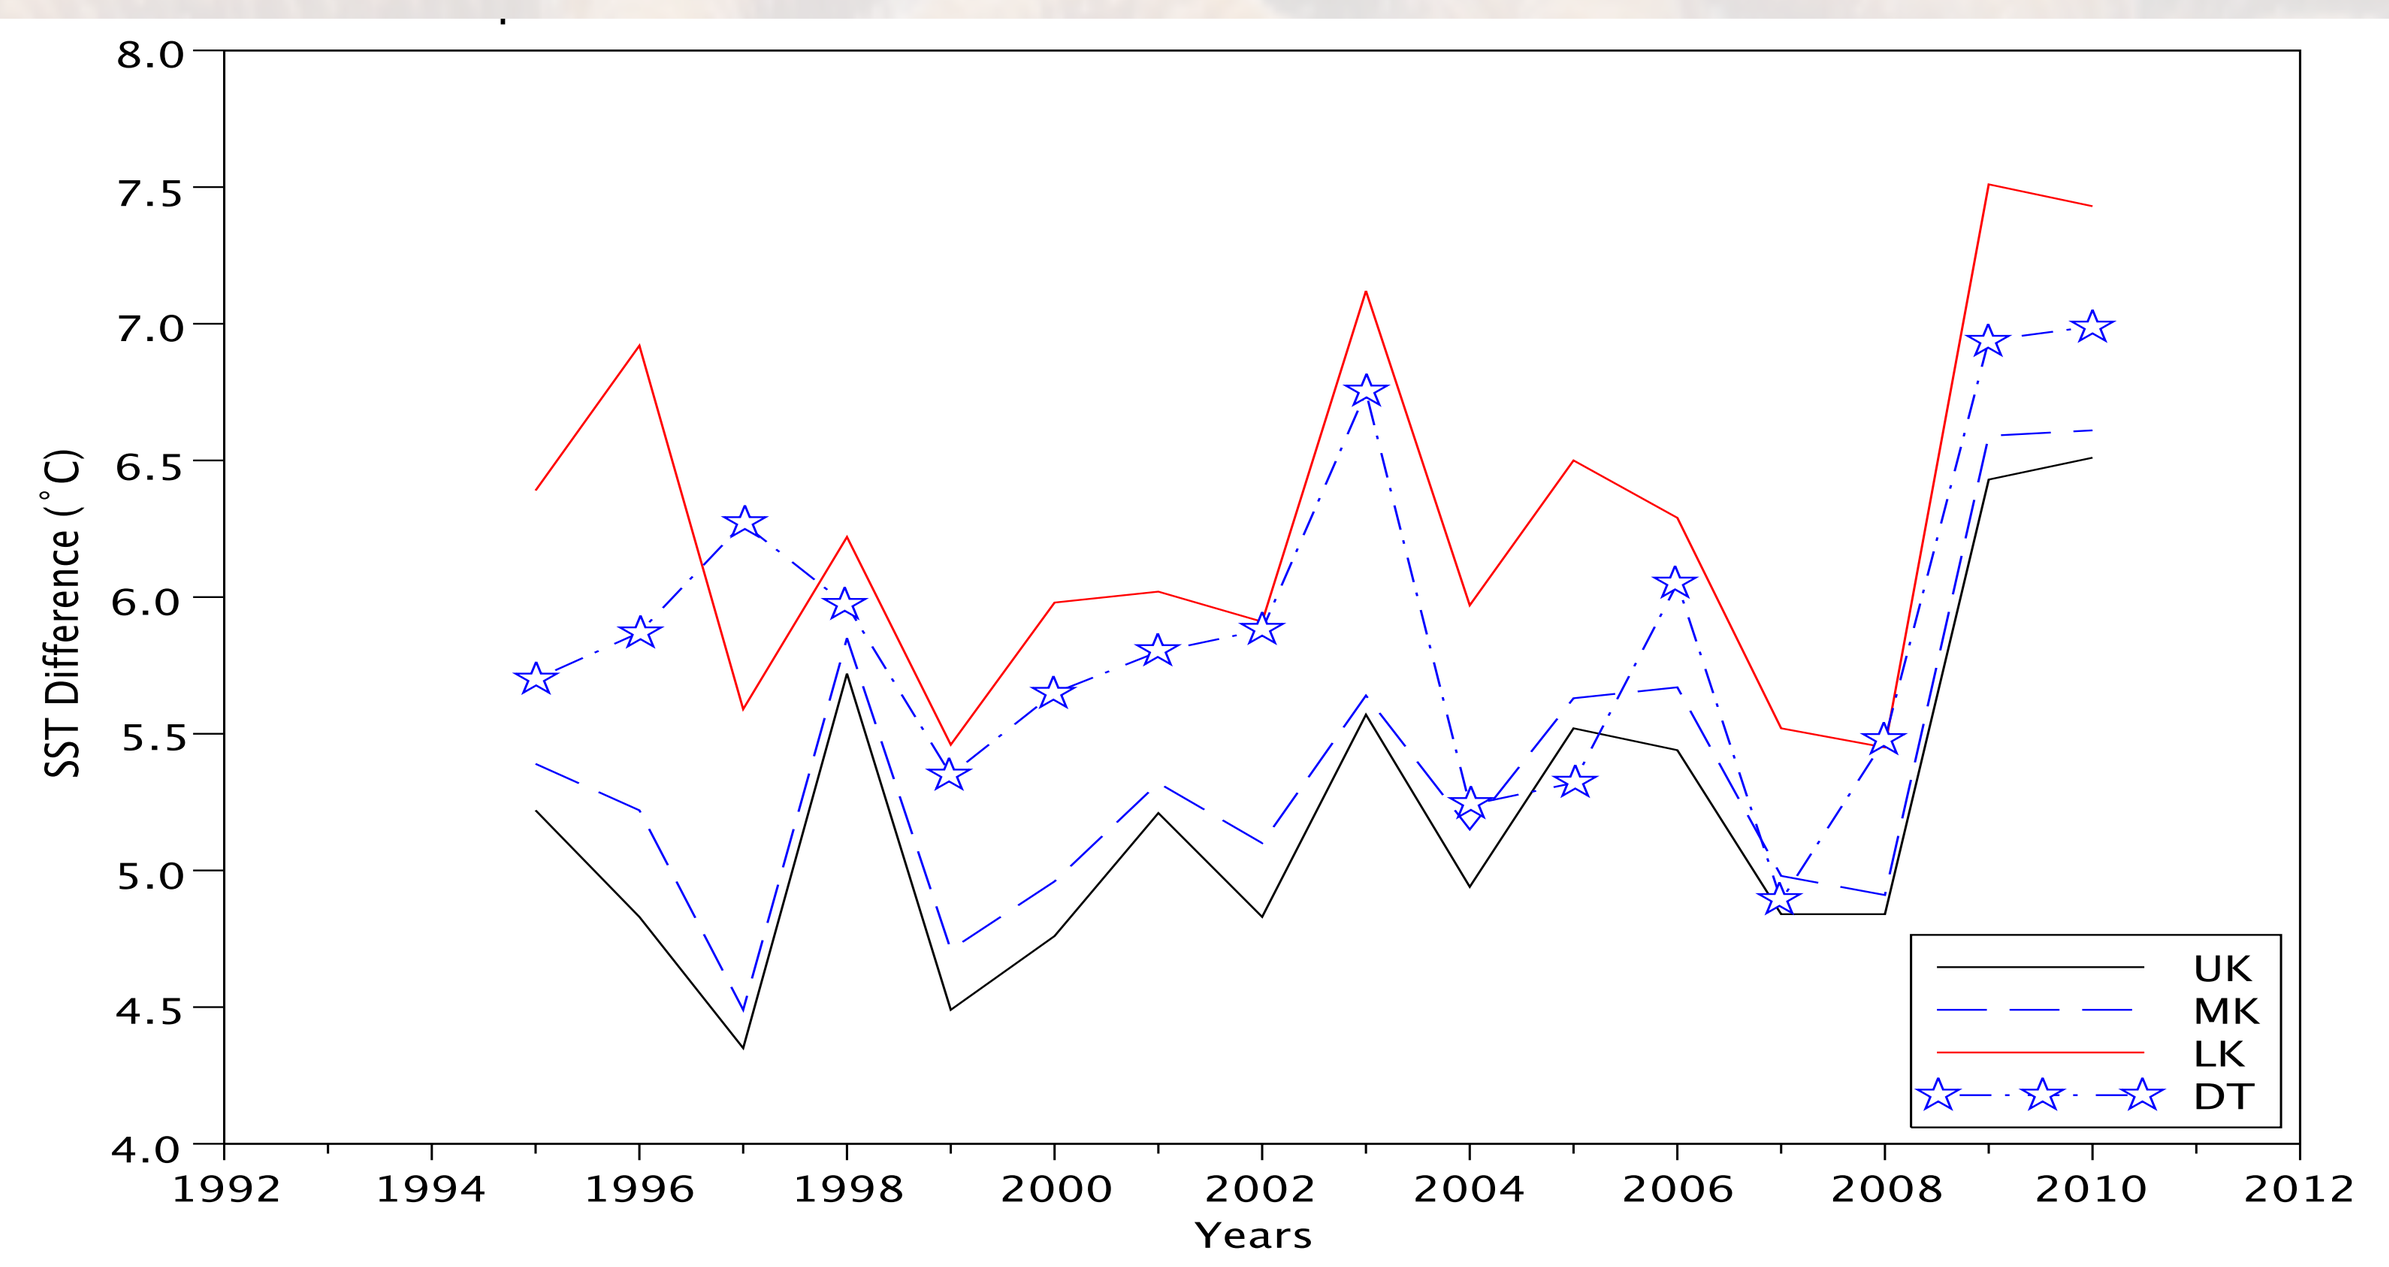

SST Difference (°C)
Years
UK
MK
LK
DT

Figure 2. Annual difference between the summer (June – October) and previous winter (December – February) AVHRR SST means for the Upper Keys (UK), Middle Keys (MK), Lower Keys (LK) and, Dry Tortugas (DT) in the Florida Keys.

- ❖ Regional maximum DHW's means were the highest for 2009-2010, exceeding the bleaching threshold of 4 in all regions (Figure 3).

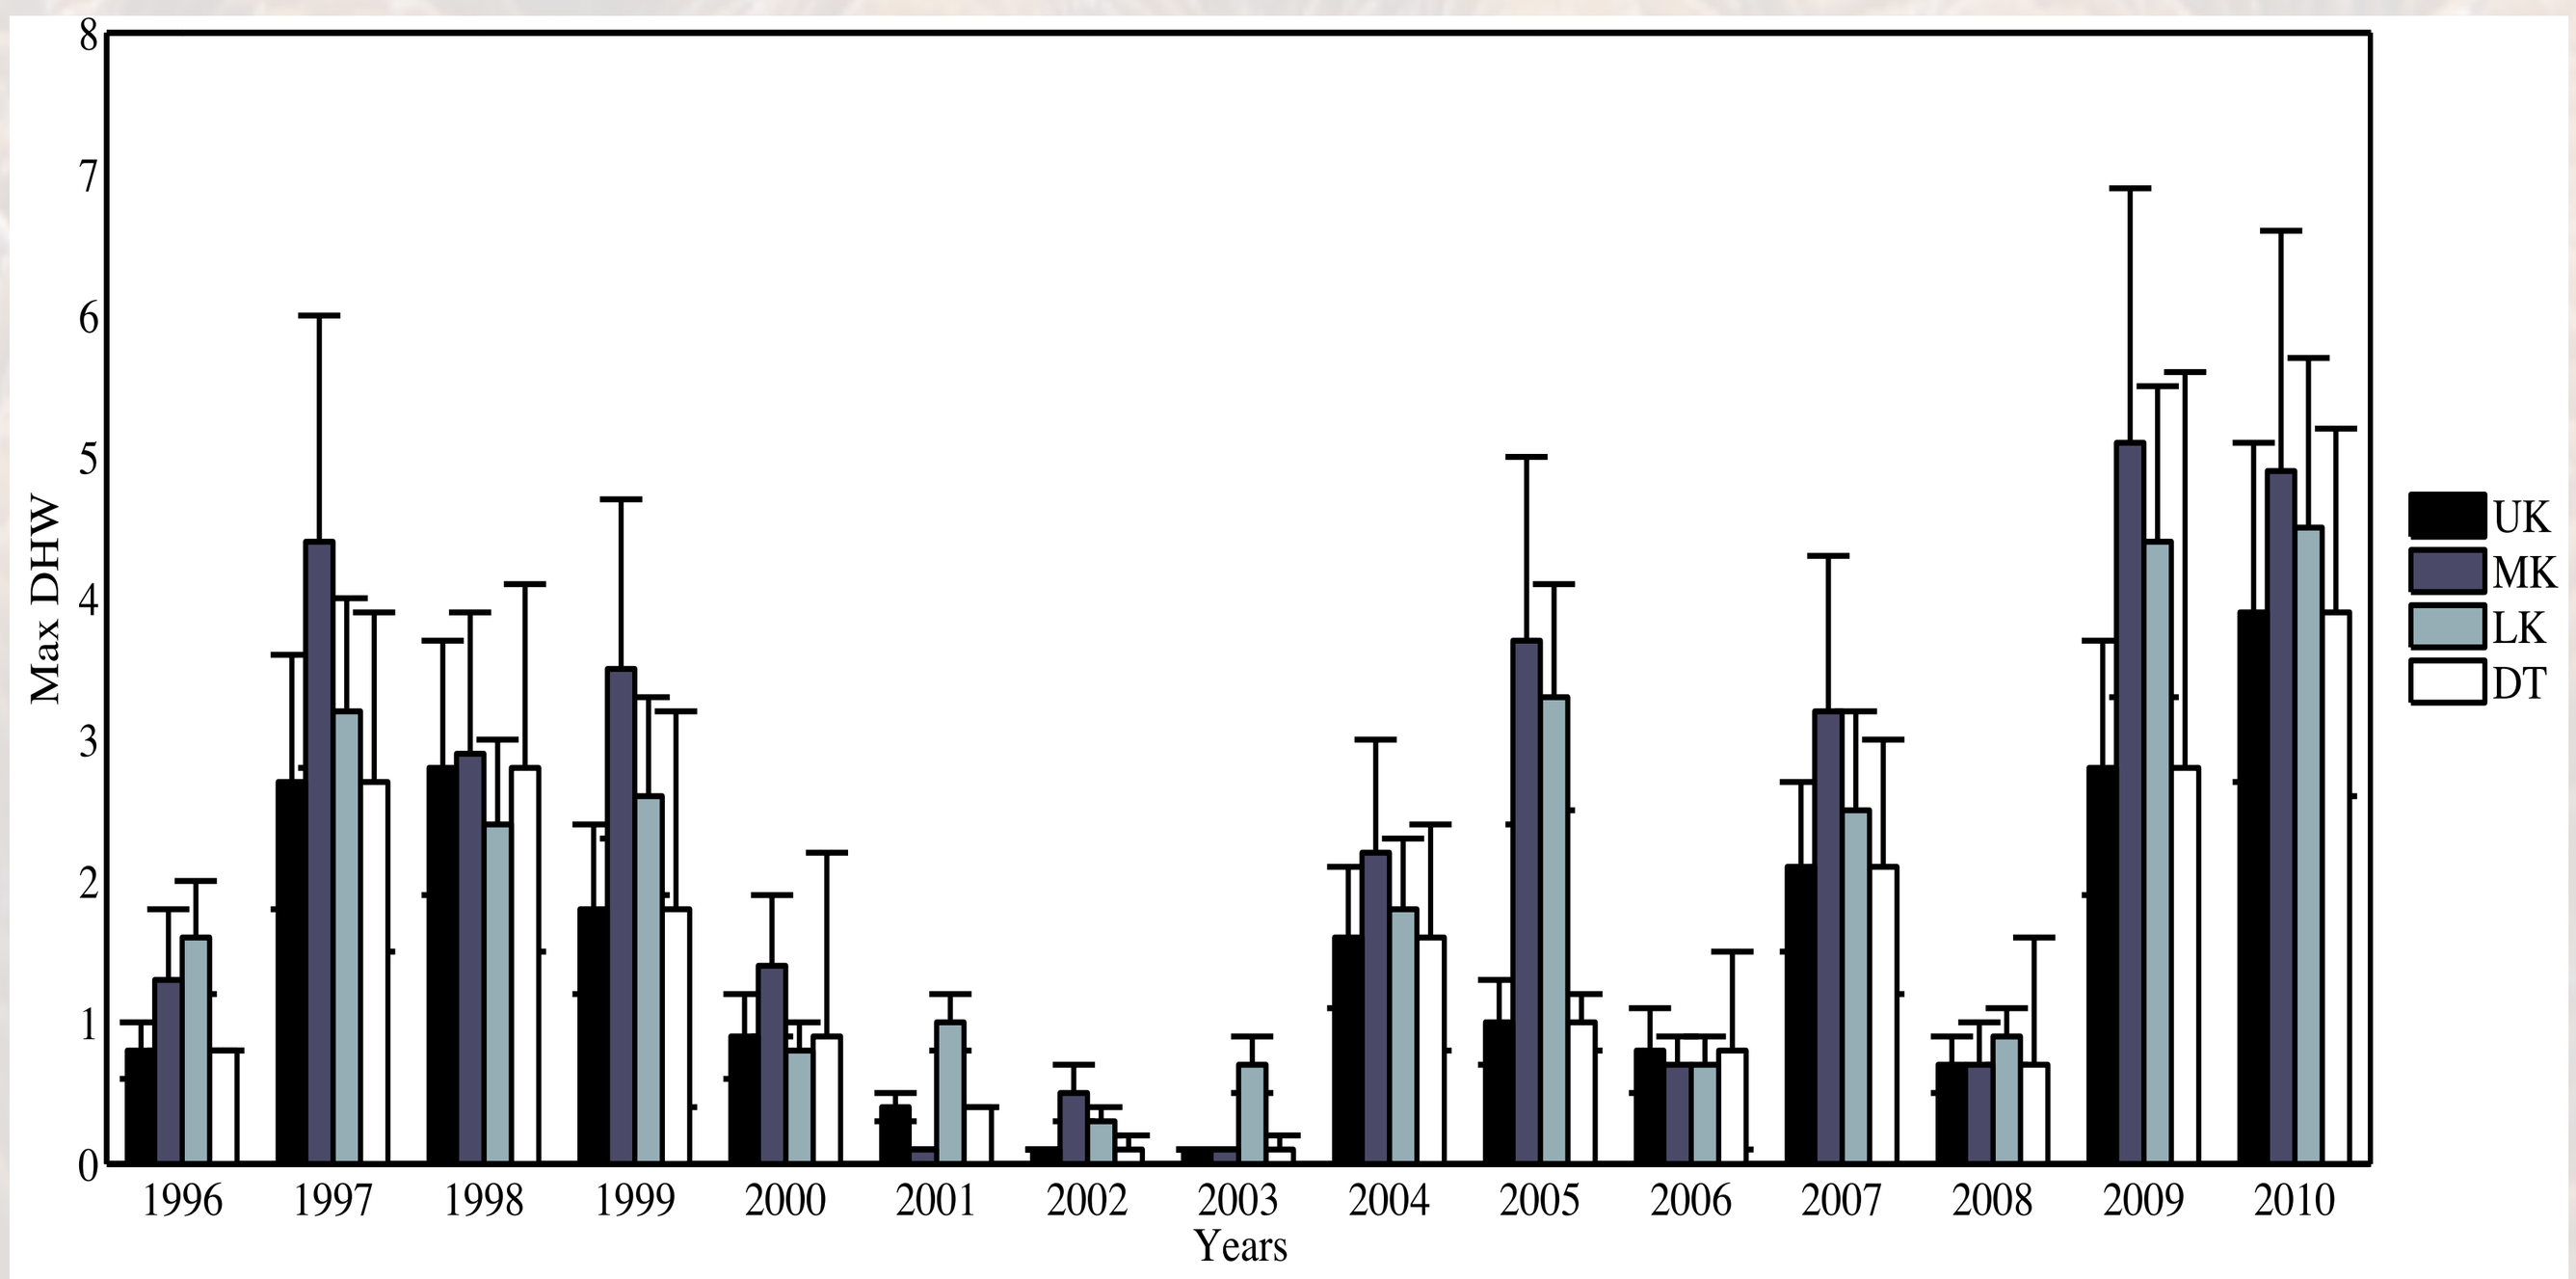

Max DHW
Years
UK
MK
LK
DT

Figure 3. Regional maximum DHW means for the Upper Keys (UK), Middle Keys (MK), Lower Keys (LK) and Dry Tortugas (DT) during 1996-2010. Standard errors are represented with one-sided error bars.

- ❖ A constrained ordination plot revealed that stony corals were strongly influenced by intermediate/high SST variability (Figure 4a). Coral species like *Montastraea cavernosa, Siderastrea siderea,* and, *Stephanocoenia intersepta* all grouped towards this SST regime (Figure 4 b).

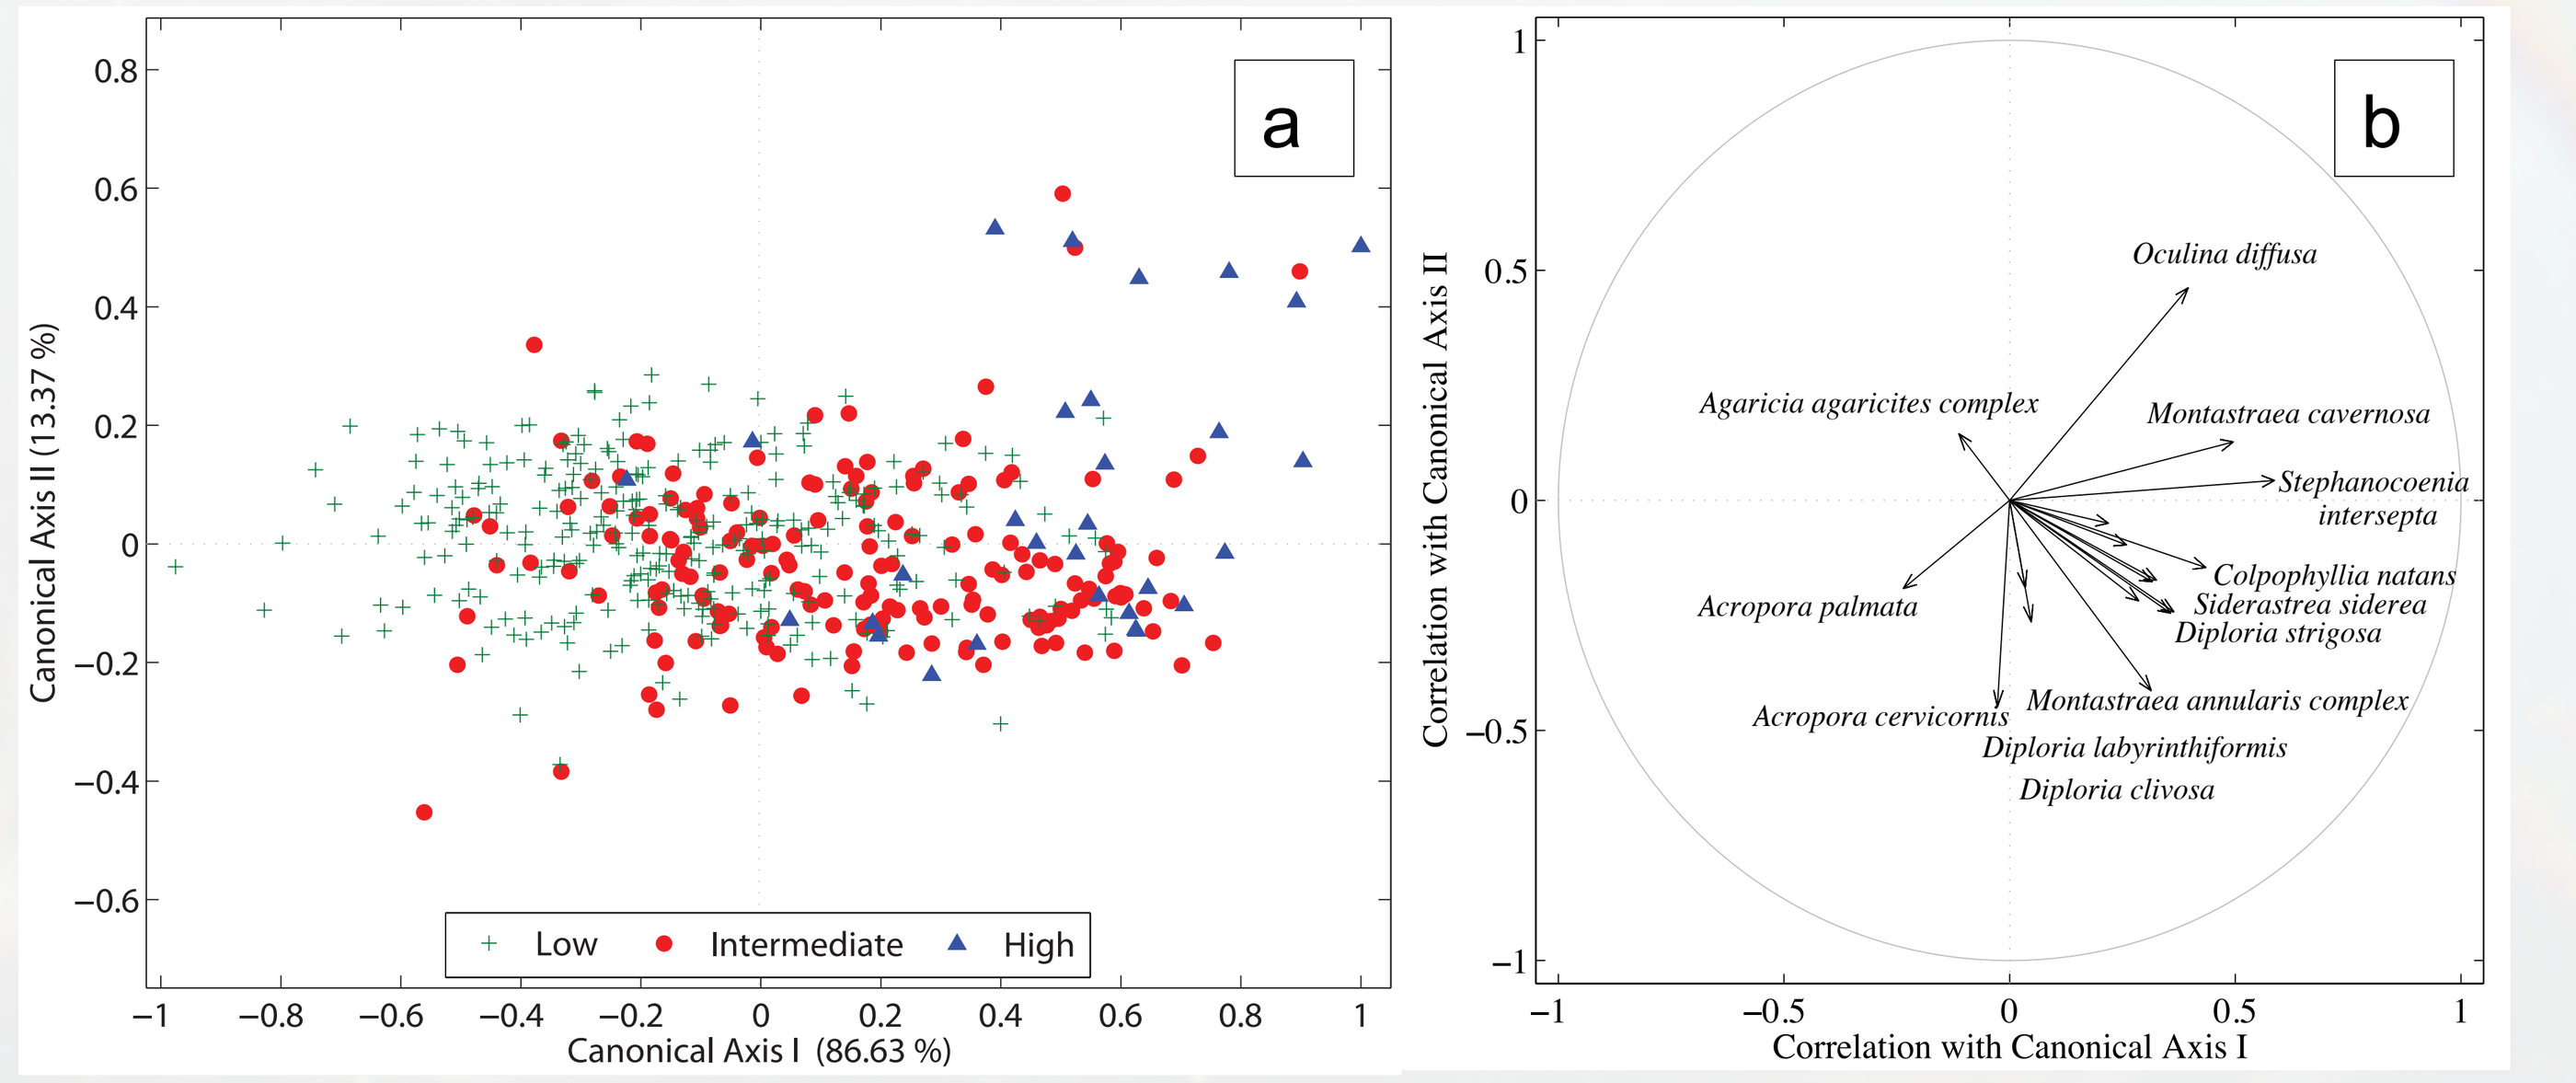

a
Canonical Axis II (13.37 %)
Canonical Axis I (86.63 %)
Low
Intermediate
High
b
Oculina diffusa
Agaricia agaricites complex
Montastraea cavernosa
Stephanocoenia intersepta
Colpophyllia natans
Siderastrea siderea
Diploria strigosa
Acropora palmata
Acropora cervicornis
Montastraea annularis complex
Diploria labyrinthiformis
Diploria clivosa
Correlation with Canonical Axis II
Correlation with Canonical Axis I

Figure 4. A: A distance-based CAP ordination plot showing the relationship of coral species percent cover among all 33 study sites in the FKNMS and the low (green cross), intermediate (red dots) and high SST variability (blue triangle) in the first two canonical axes. **B:** Ordination bi-plot of the six most important species for each axis.

- ❖ Results reveal that, after the thermal anomalies conducive to bleaching events in 1997-1998, increases in the maximum DHW's led to significant increases in H' for all regions (Figure 5a). However, a decrease in yearly rate of change for H' was observed in the Middle Keys for years 2009-10 (Figure 5b).

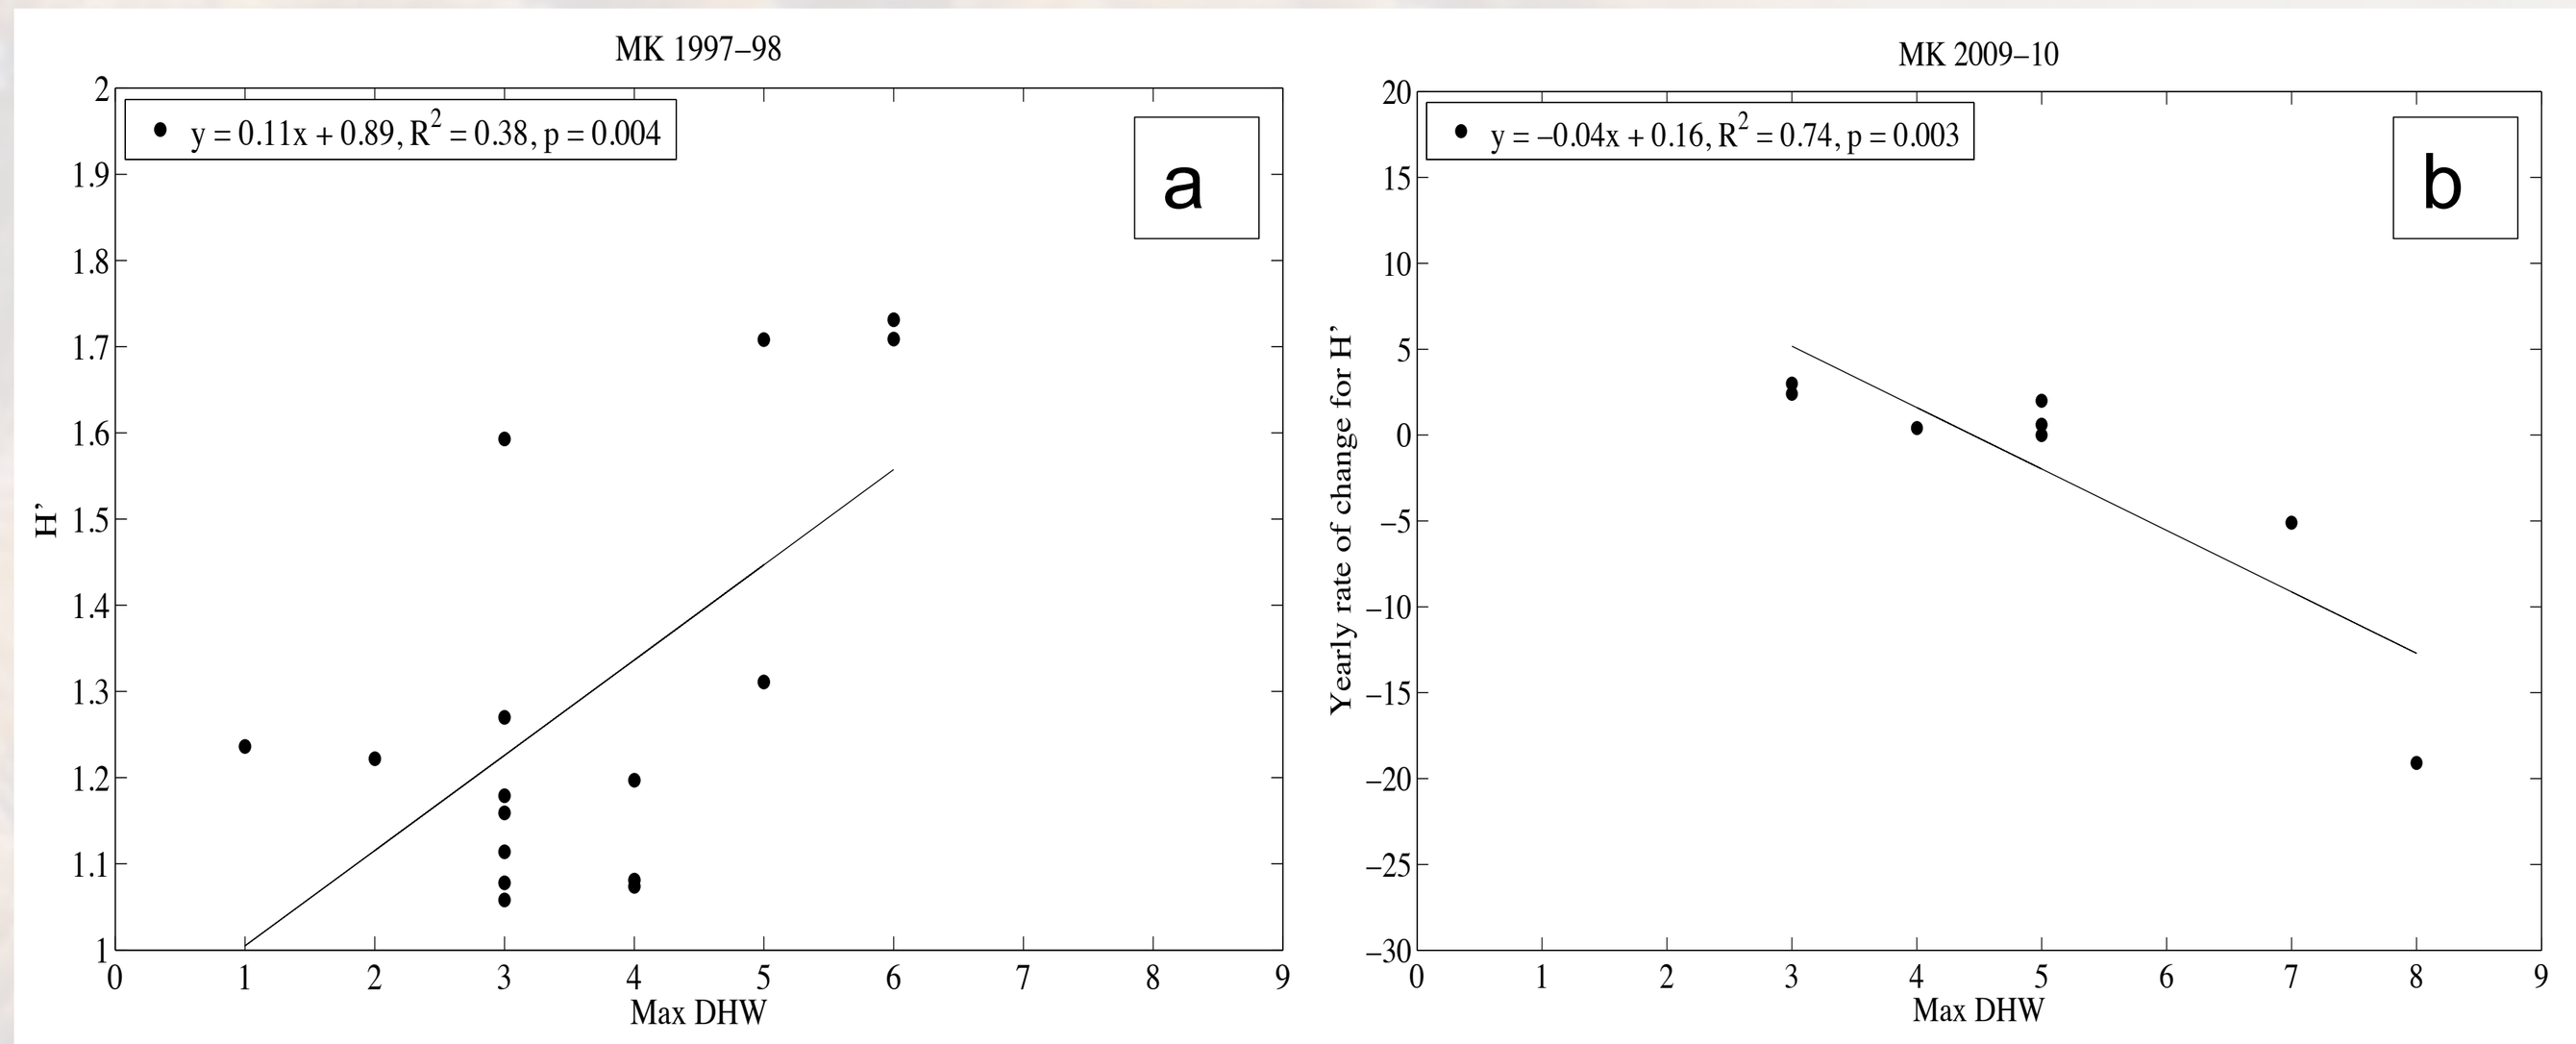

MK 1997–98
y = 0.11x + 0.89, R² = 0.38, p = 0.004
a
H'
Max DHW
MK 2009–10
y = −0.04x + 0.16, R² = 0.74, p = 0.003
b
Yearly rate of change for H'
Max DHW

Figure 5. A: Regional maximum Degree Heating Weeks (DHW's) means compared to the Shannon diversity index (H') for 1998-99 in the Middle Keys. **B:** Regional maximum DHW's means for years 2009-2010 compared to the yearly rate of change (2009-10) of H' in the Middle Keys. Significance is at p-value < 0.05.

Preliminary Conclusions

- ❖ Stony corals were strongly influenced by intermediate (7 – 10.9 °C) /high (≥ 11 °C) SST variability.
- ❖ Despite exposure to three bleaching events since 1996, reefs in the Middle Keys experienced significant losses in diversity due to warm thermal anomalies only in 2009-2010.

Work In Progress

- ❖ Assessing the utility of the MODIS & AVHRR thermal indices in the context of coral reef bleaching surveys after cold and warm events in the Florida Keys.

Acknowledgements

Funding for this study was provided by: NASA Applications Program /Ecological Forecasting Application Area, NOAA NESDIS Coral Reef Watch, UNEP WCMC, NASA Earth and Science 2012/2013 Fellowship, University of South Florida (USF), USF – FGLSAMP Bridge to the Doctorate Program

[1] University of South Florida - College of Marine Science, [2] NOAA Coral Reef Watch, [3] NASA Ames Research Center, [4] University of Colorado – Cooperative Institute for Research in Environmental Sciences (CIRES), [5] Fish & Wildlife Research Institute – Coral Reef Evaluation and Monitoring Project (CREMP)